EVO756 inhibits activation of MRGPRX2 *in vitro* and correlates to human data

Jamie L. Harden[1*], Sreya Bagchi[1], Stefan Frischbutter[2,3], Jorg Scheffel[2,3], Han Gao[2,3], Nana Shi [2,3], Alexandra Pavel[1], Polina Bulkspun[1], Andrea Kim[1], Jeegar Patel[1], Lorena Riol-Blanco[1]

[1]Evommune, Inc. Palo Alto, CA 94304, [2]Institute of Allergology, Charité – Universitätsmedizin Berlin, Corporate Member of Freie Universität Berlin and Humboldt-Universität zu Berlin, Berlin, Germany
*[3]Fraunhofer Institute for Translational Medicine and Pharmacology ITMP, Immunology and Allergology IA, Berlin, Germany *presenting author*

Background

EVO756 is a novel, small molecule antagonist of the Mas-related G protein-coupled receptor X2 (MRGPRX2), a receptor found on mast cells, causing IgE-independent degranulation, and on sensory neurons associated with itch, pain and inflammation.

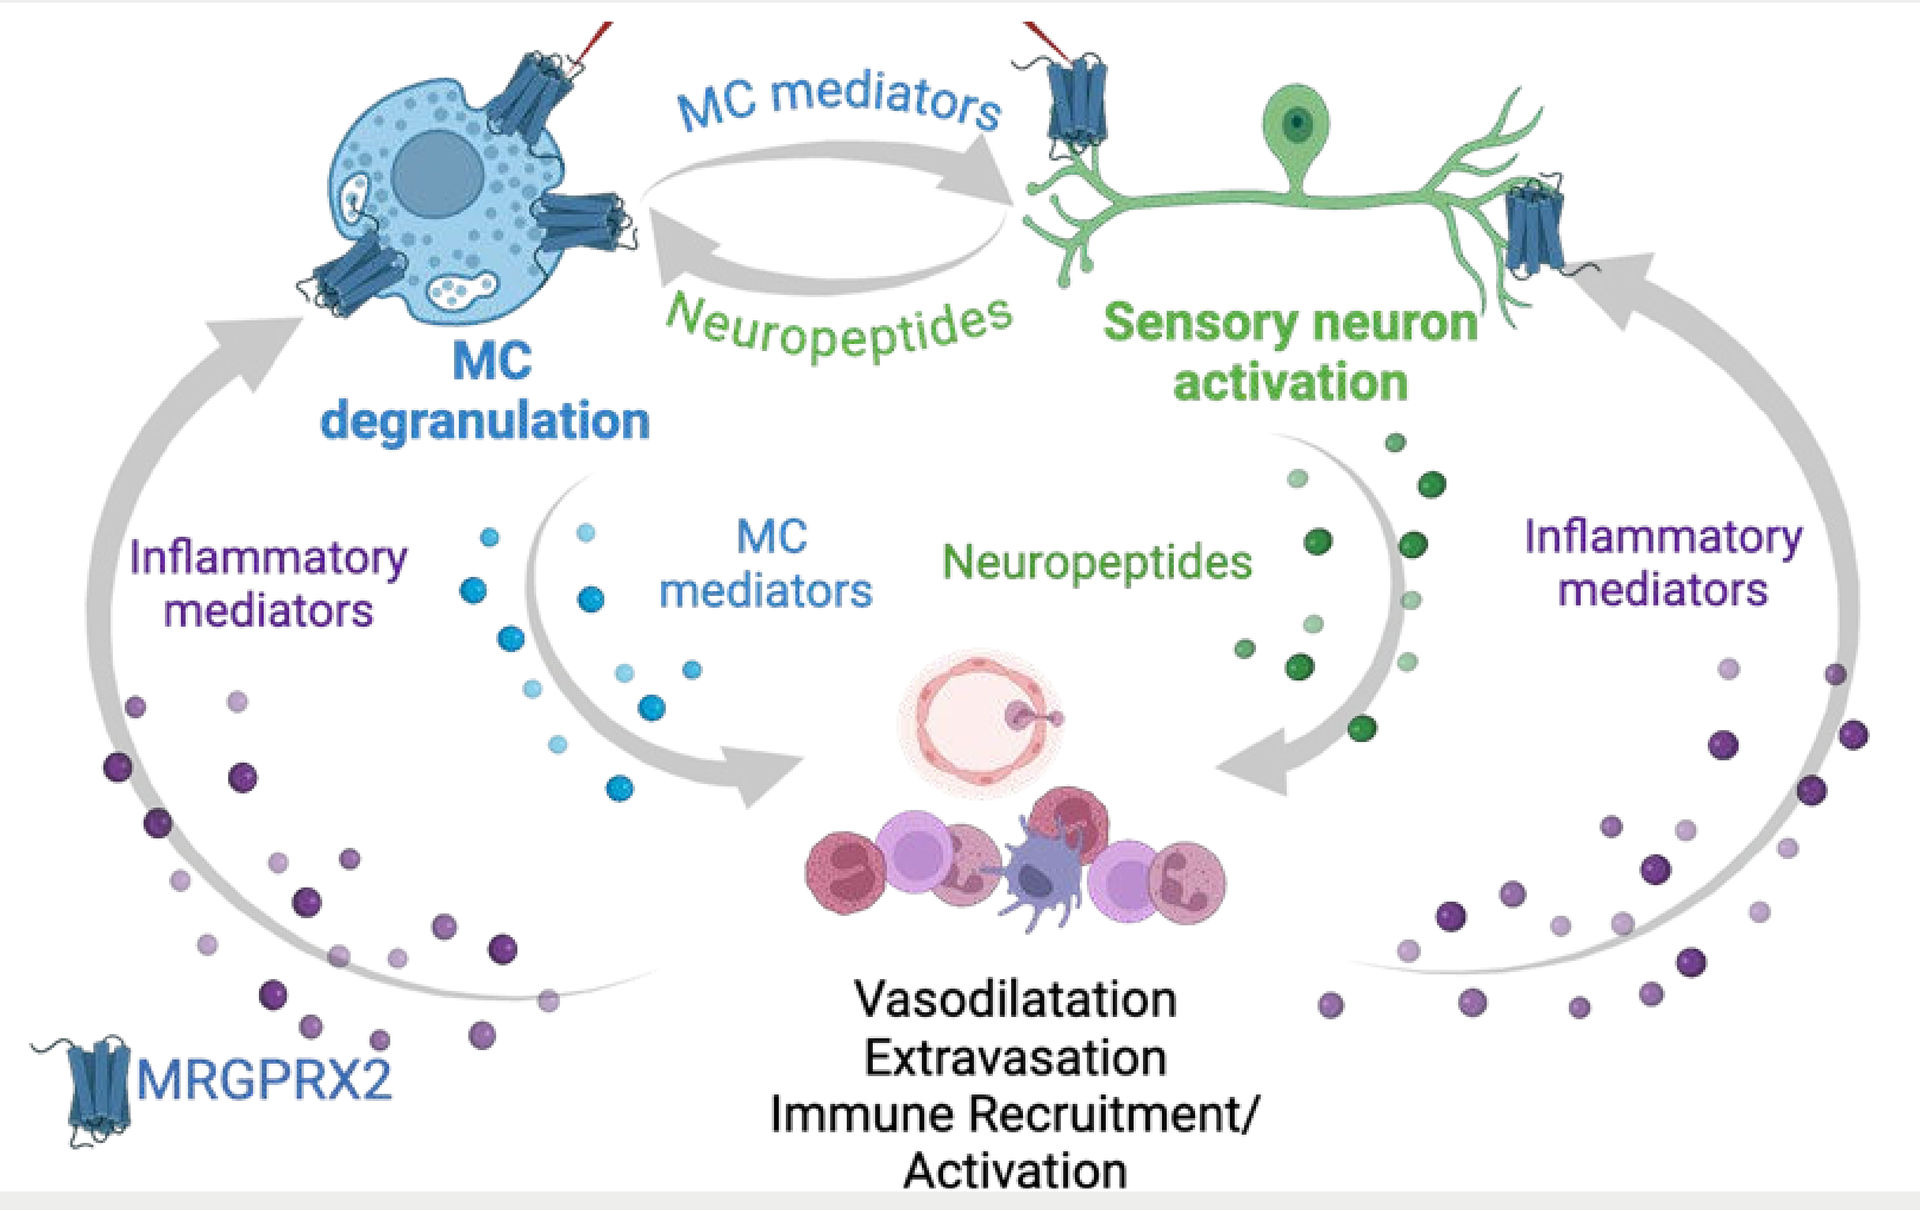
MC mediators
Neuropeptides
MC degranulation
Sensory neuron activation
Inflammatory mediators
MC mediators
Neuropeptides
Inflammatory mediators
MRGPRX2
Vasodilatation
Extravasation
Immune Recruitment/
Activation

Selectively modulating MRGPRX2 represents a novel therapeutic approach for mast cell-mediated diseases. MRGPRX2 is a promiscuous receptor that is responsive to a multitude of endogenous and exogenous ligands. One exogenous MRGPRX2 ligand is the FDA-approved bradykinin-B2 receptor antagonist, icatibant.

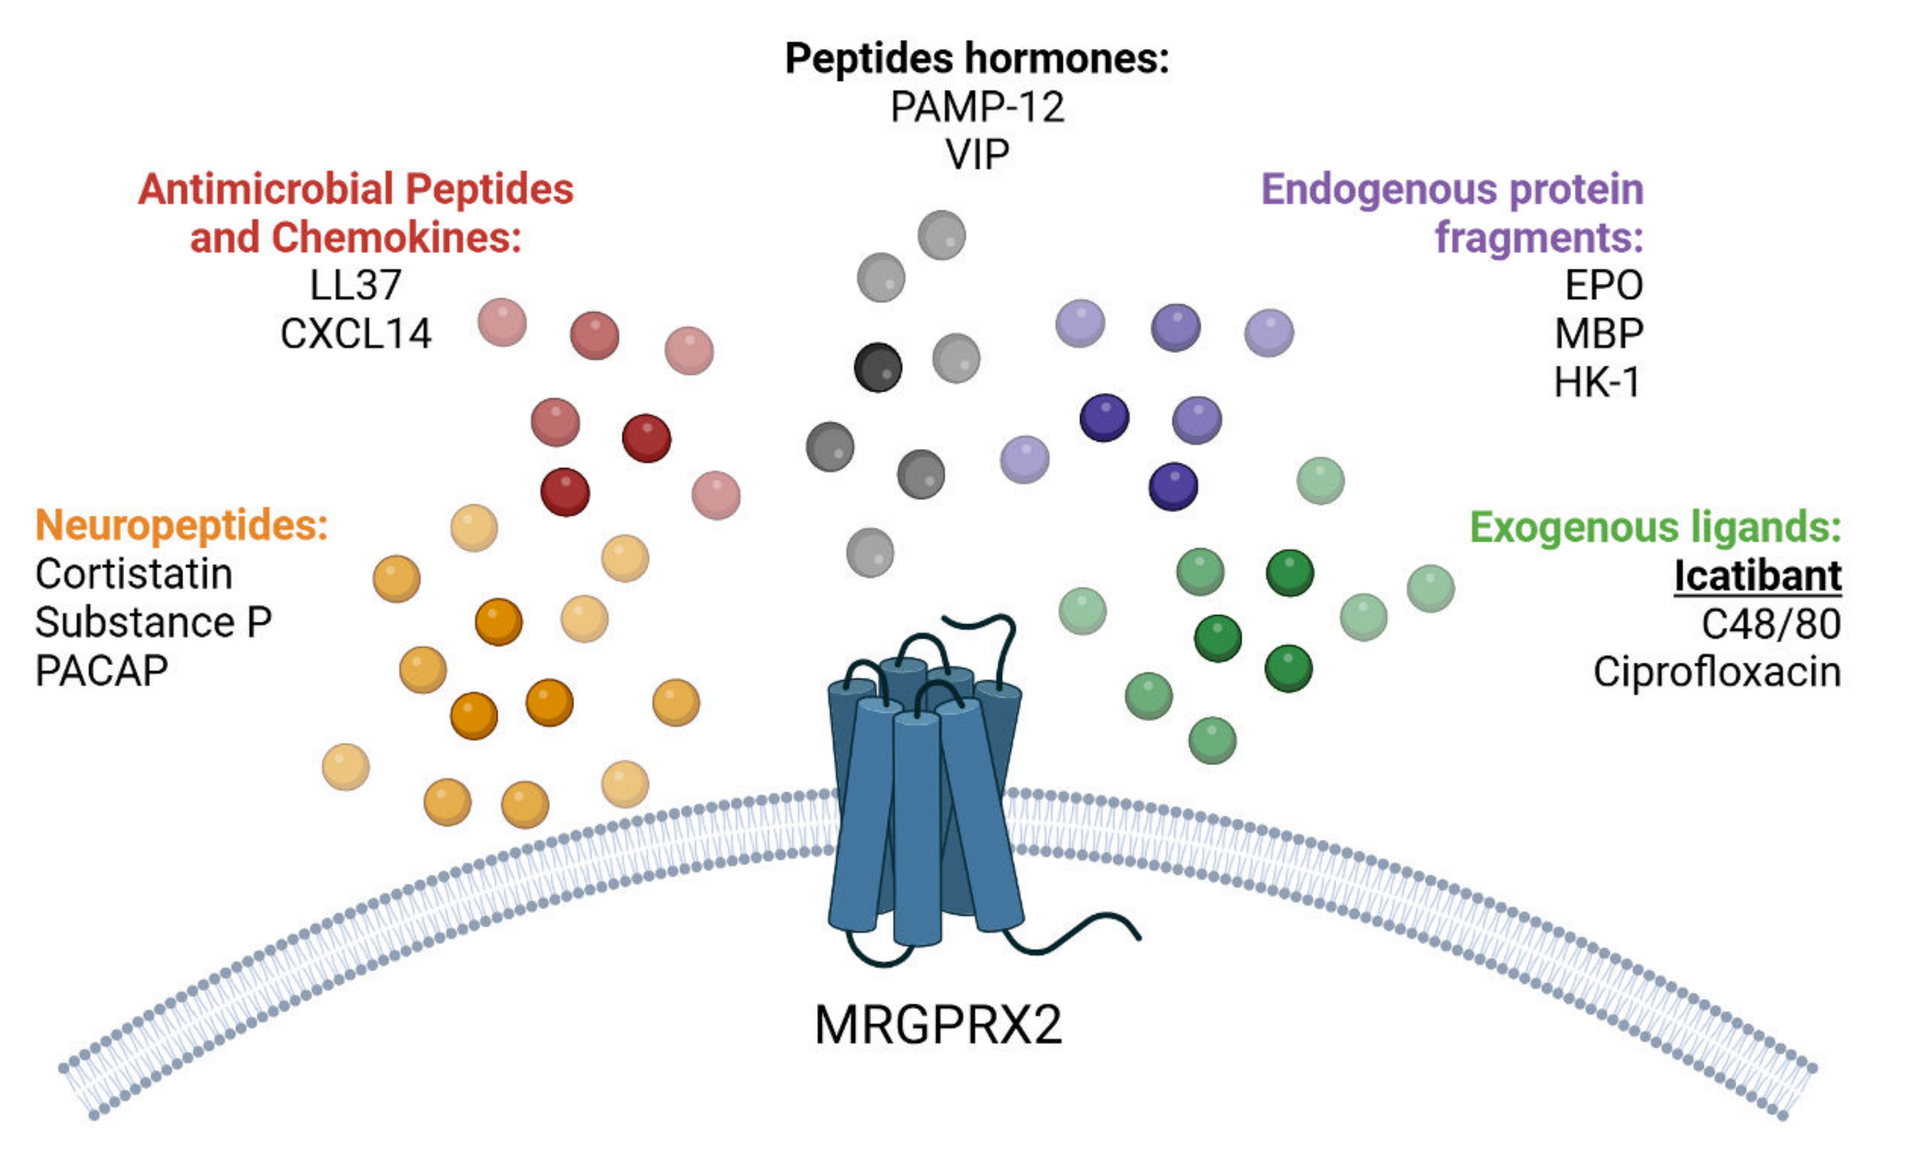
Peptides hormones:
PAMP-12
VIP
Antimicrobial Peptides and Chemokines:
LL37
CXCL14
Endogenous protein fragments:
EPO
MBP
HK-1
Neuropeptides:
Cortistatin
Substance P
PACAP
Exogenous ligands:
Icatibant
C48/80
Ciprofloxacin
MRGPRX2

Previous work has demonstrated that an intradermal icatibant injection into healthy subjects forms a measurable wheal response (*Shtessel, M. et al., J Invest Derm (2021) 141,678e681*). Here, we characterized responses to icatibant versus endogenous MRGPRX2 ligands, as well as characterized the ability of EVO756 to inhibit this response, in multiple *in vitro* models. The aim of this work is to demonstrate that icatibant can be considered a prototypical MRGPRX2 ligand and supports the utility of using icatibant to determine MRGPRX2 target engagement in a Phase 1 healthy volunteer skin challenge study with oral EVO756.

Methods

hMRGPRX2-CHO assay – *hMRGPRX2 agonists at EC80 were used to evaluate the ability of EVO756 to inhibit calcium flux (FLIPR® assay) in hMRGPRX2-CHO cells, data plotted as % of max response of the hMRGPRX2-CHO cells to vehicle + agonist.*

Primary human skin MCs (phsMCs) *were isolated from human skin obtained from plastic surgeries under an IRB approved protocol and informed consent. Skin biopsies were incubated overnight in dispase II and the epidermis separated from the dermis. The dermis was finally chopped and then was incubated with collagenase, hyaluronidase and DNaseI. Anti-CD117 antibody conjugated magnetic beads were used to obtain a pure population of phsMCs.*

Mast cell degranulation assay – *phsMCs were pretreated for a maximum of 5 minutes with EVO756 prior to stimulation with MRGPRX2 agonists at $EC_{80\text{-}100}$ concentrations. Degranulation was evaluated by cell surface expression of CD63 or CD107a via flow cytometry, or beta-hexosaminindase release.*

Spring Science platform (morphology and time-course degranulation) - *phsMCs isolated as described above were rested overnight, and the next day stained with Hoechst, LysoTracker Deep Red and CellBrite NIR 750. Cells were then washed and stained with an antibody against CD63, prior to stimulation with MRGPRX2 agonists. Cells were then imaged on an ImageXpress Micro Confocal High-Content Imaging System in widefield on a 40x ELWD air objective. Morphological changes were characterized based on brightfield images, Hoechst and CellBrite stains, while CD63 expression was characterized by expression of the marker on the cell surface detected by the imaging system.*

RNA sequencing – *Primary human skin mast cells were cultured with the EC_{100} of various MRGPRX2 agonists for 4 hours. Mast cells were then collected, lysed and RNA extracted using a Qiagen Rneasy micro kit. RNA was sent to MedGenome for library preparation and sequencing.*

***Ex vivo* skin stimulation** – *whole human skin was injected with 20 ul of a 10 uM solution of EVO756 immediately prior to injection at the same site with 20ul of codeine (0.045%) or icatibant (100, 250, or 500 ug/ml). The PELSA system (Ascilion Inc.), which involves hollow microneedles and mild vacuum, were utilized to extract dermal interstitial fluid (dISF) from the injection sites, and histamine levels were quantified.*

Results

Icatibant induces similar responses in primary human MCs compared to endogenous MRGPRX2 ligands

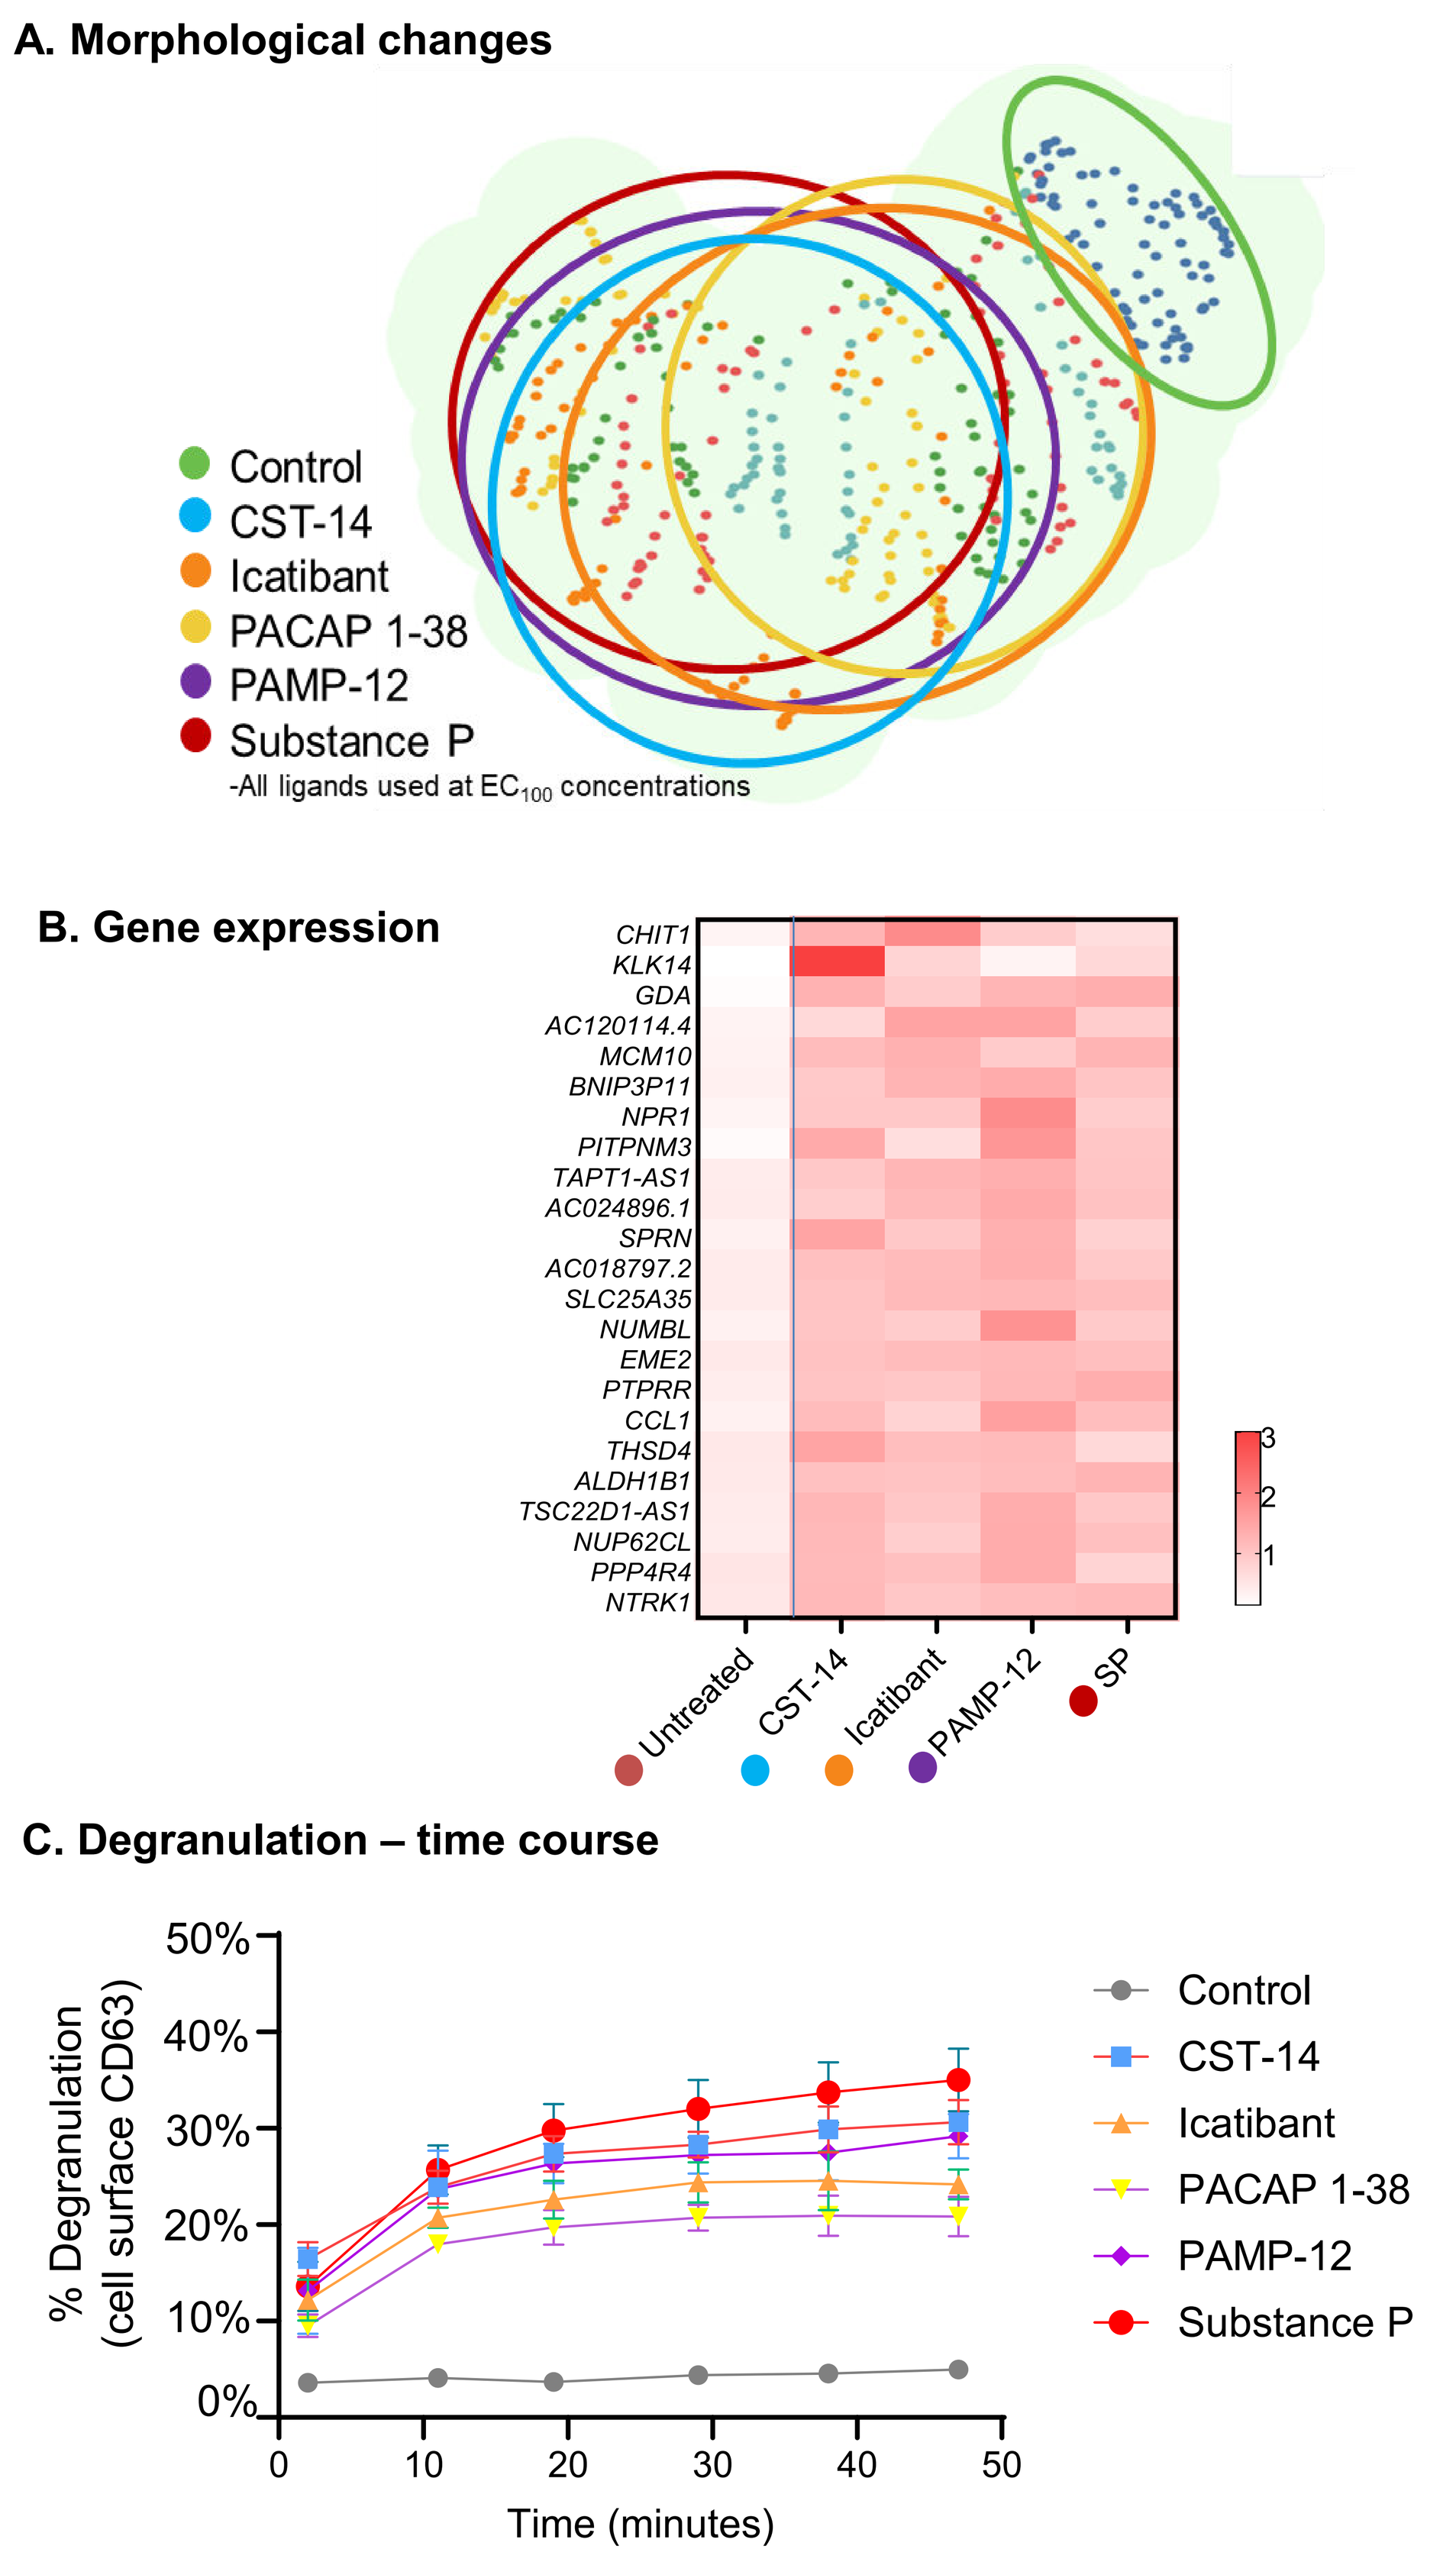
A. Morphological changes
Control
CST-14
Icatibant
PACAP 1-38
PAMP-12
Substance P
-All ligands used at EC_{100} concentrations
B. Gene expression
CHIT1
KLK14
GDA
AC120114.4
MCM10
BNIP3P11
NPR1
PITPNM3
TAPT1-AS1
AC024896.1
SPRN
AC018797.2
SLC25A35
NUMBL
EME2
PTPRR
CCL1
THSD4
ALDH1B1
TSC22D1-AS1
NUP62CL
PPP4R4
NTRK1
Untreated
CST-14
Icatibant
PAMP-12
SP
C. Degranulation – time course
% Degranulation (cell surface CD63)
Time (minutes)
Control
CST-14
Icatibant
PACAP 1-38
PAMP-12
Substance P

Figure 1. (A) PCA of morphology of primary human skin mast cells stimulated with MRPGRX2 agonists versus unstimulated control cells (B) Common differentially expressed genes in phsMCs stimulated with MRGPRX2 agonists versus untreated cells (C) Time-course of phsMC degranulation upon stimulation with MRGPRX2 agonists, as determined by the percent of phsMCs with cell-surface CD63 staining

EVO756 inhibits icatibant induced activation of MRGPRX2, similarly to endogenous ligands

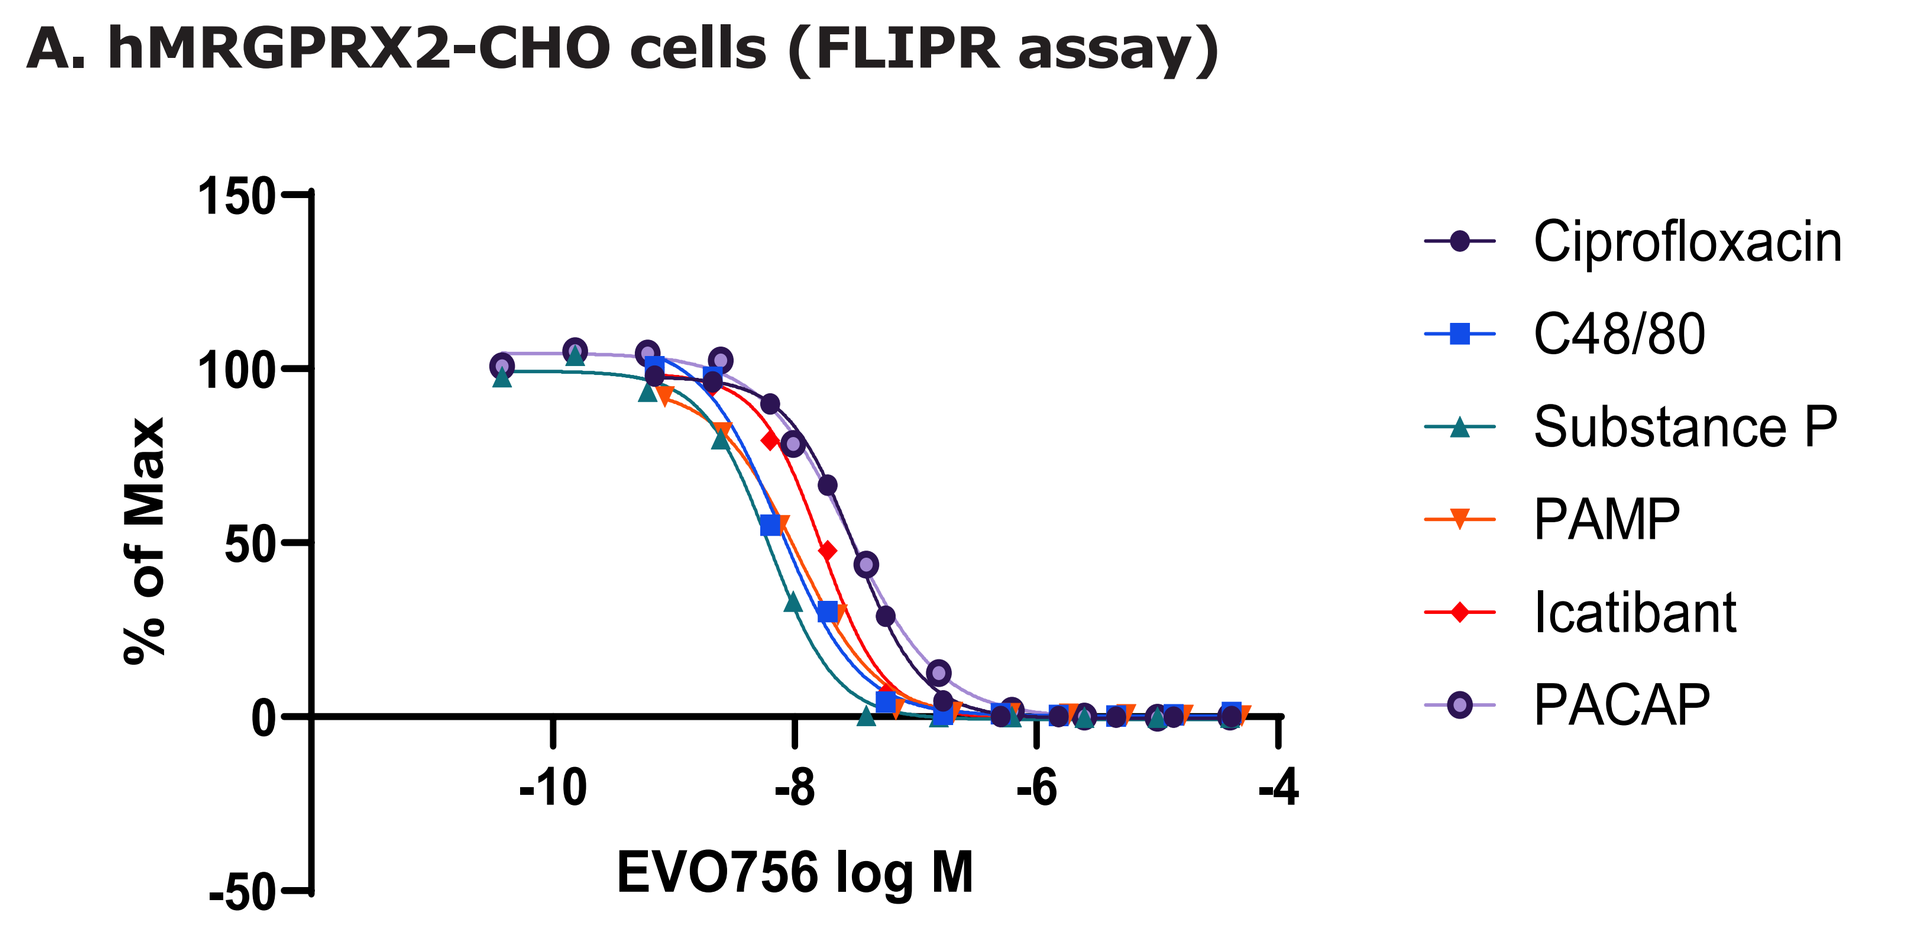
A. hMRGPRX2-CHO cells (FLIPR assay)
% of Max
EVO756 log M
Ciprofloxacin
C48/80
Substance P
PAMP
Icatibant
PACAP

Endogenous	EVO756 IC_{50} (nM)	
Neuropeptides	Substance P	5.3
	PACAP 1-38	30.2
Hormones	PAMP-12	9.2
Exogenous		
Drugs	**Icatibant**	**12.9**
Antibiotics	Ciprofloxacin	16.6
Other	C48/80	6.7

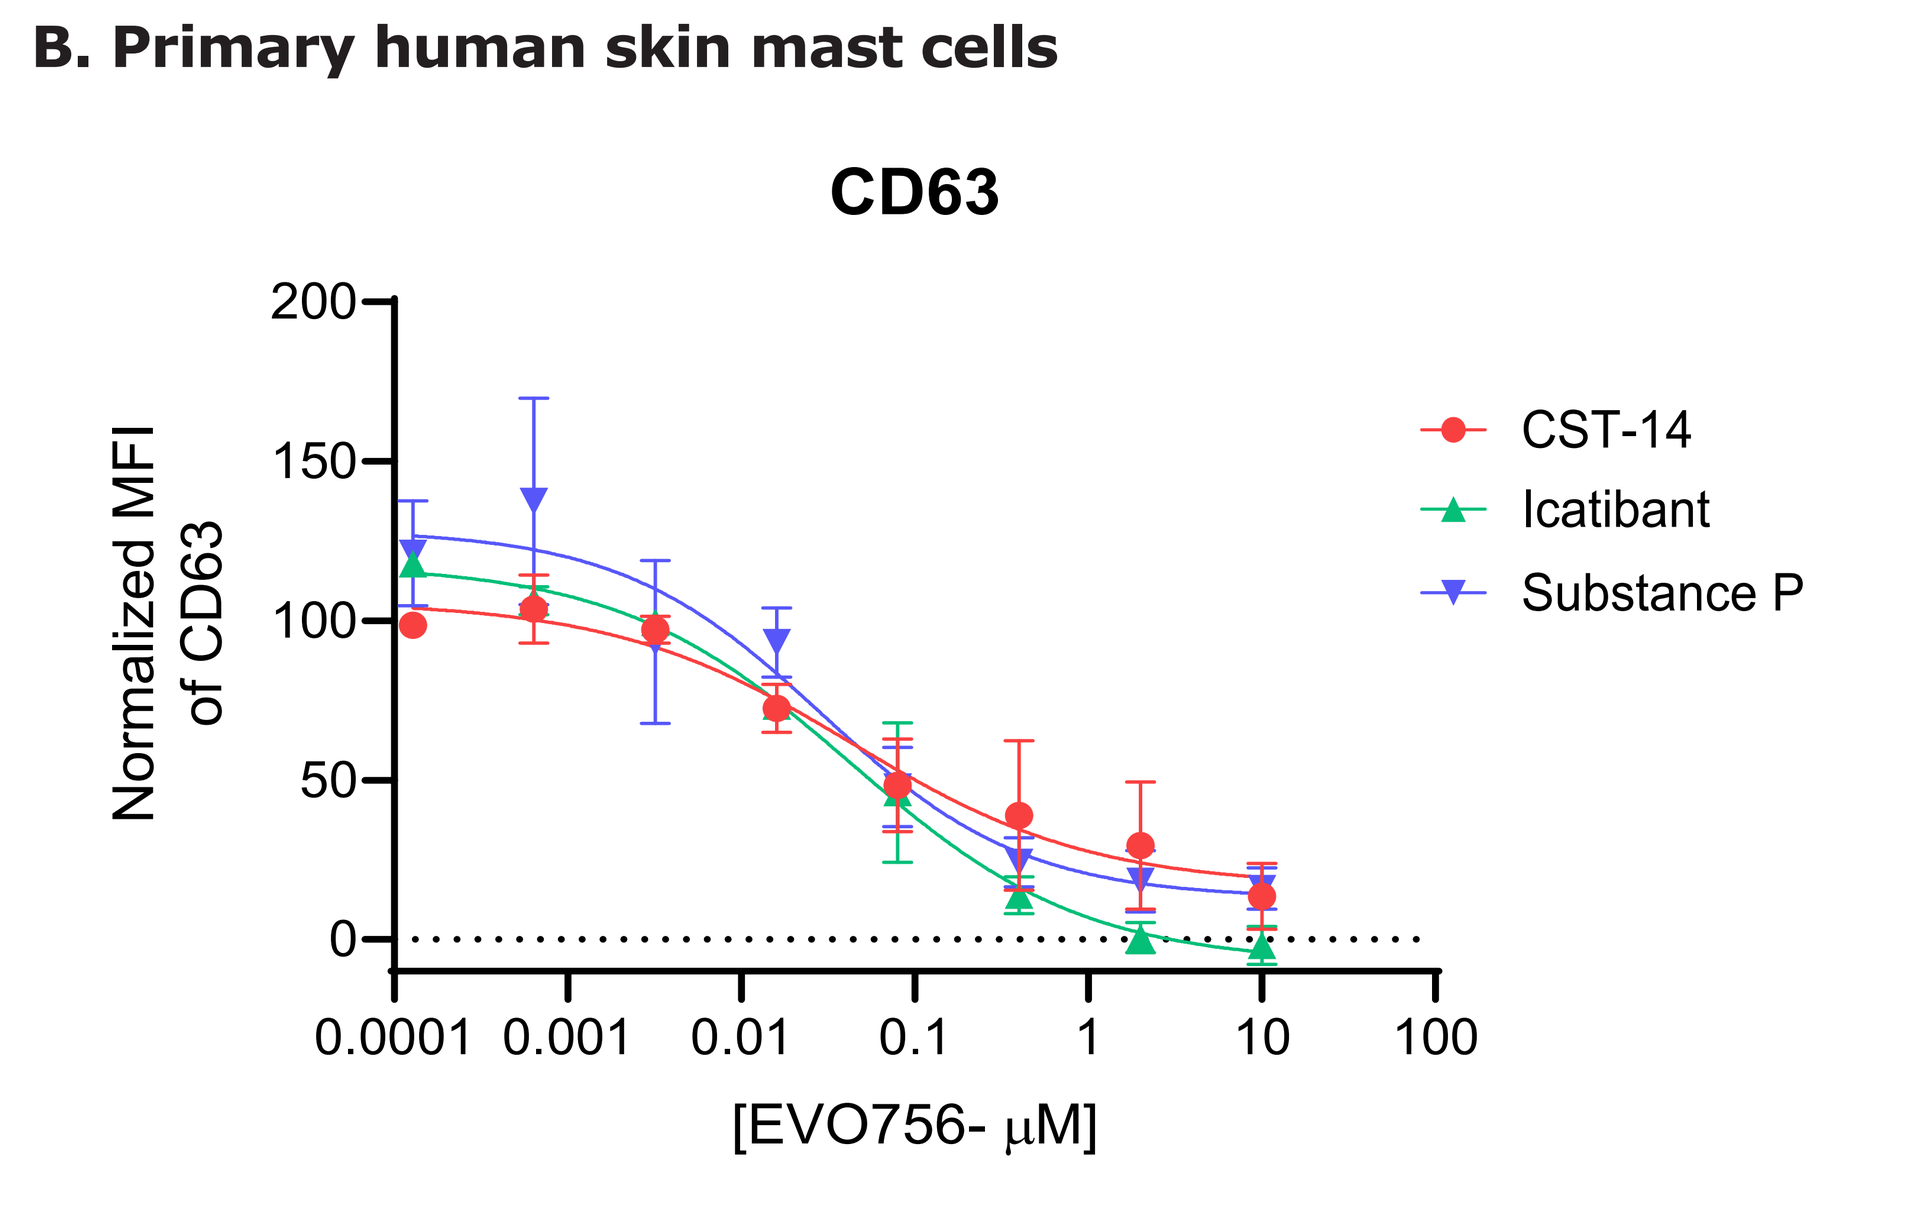
B. Primary human skin mast cells
CD63
Normalized MFI of CD63
[EVO756- μM]
CST-14
Icatibant
Substance P

	EVO756 IC_{50} (nM)		
	CD63	CD107a	B-hex
Cortistatin-14	44	67	82
Substance P	29	27	63
Icatibant	**43**	**95**	**51**

Figure 2. (A) EVO756 concentration-dependent inhibition of hMRGPRX2-CHO cell calcium mobilization induced by MRGPRX2 agonists and calculated IC50 values for EVO756 (B) EVO756 concentration-dependent inhibition of degranulation in phsMC, as determined via cell surface CD63 expression via flow cytometry (similar data was found for CD107a cell surface expression and beta-hexosaminindase release) and EVO756 IC50 values

Results *(cont.)*

EVO756 inhibits icatibant and codeine induced histamine release in an *ex vivo* human skin assay

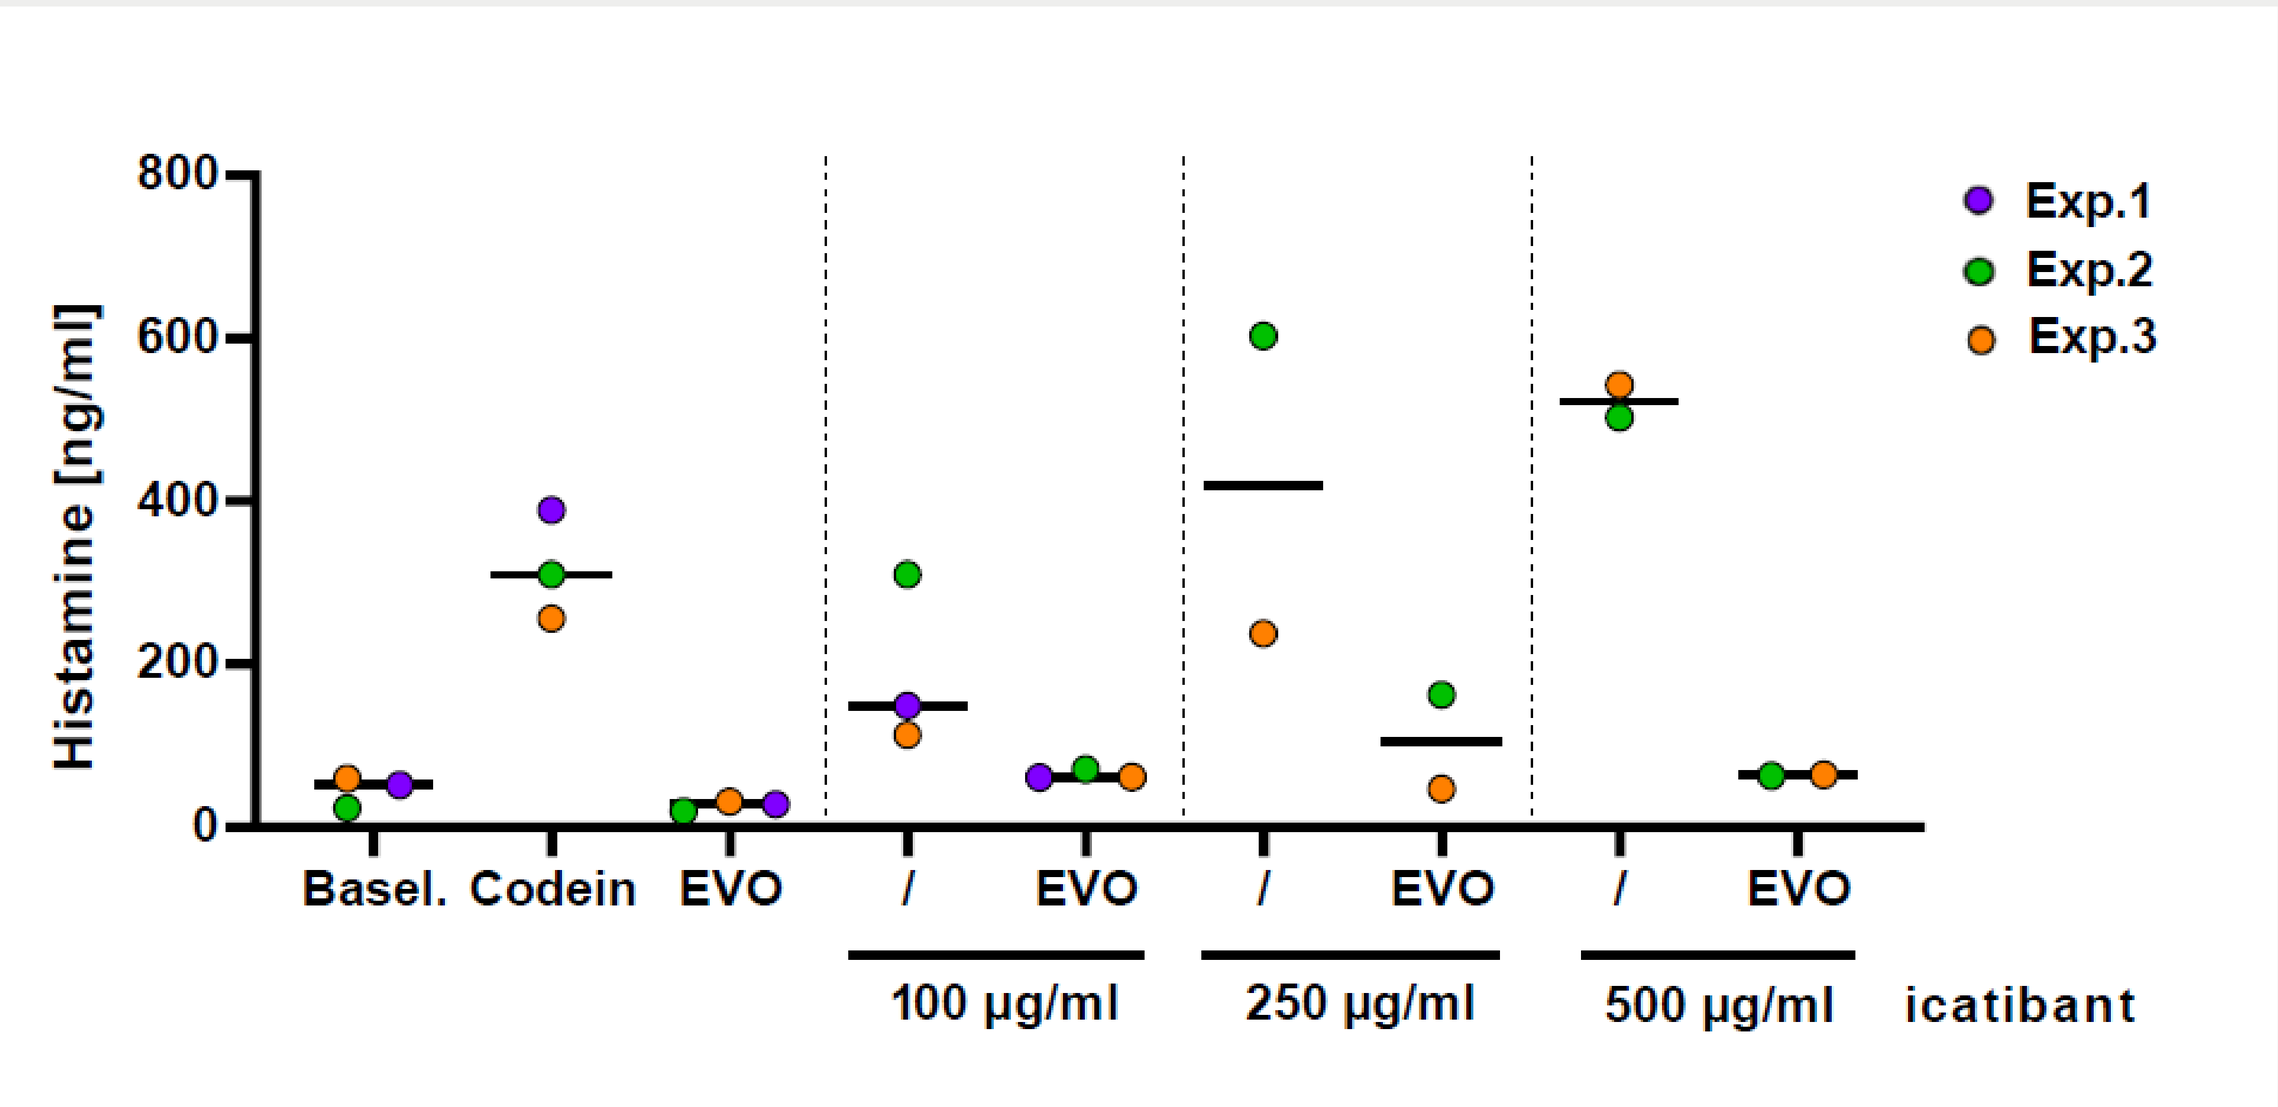
Histamine [ng/ml]
Exp.1
Exp.2
Exp.3
Basel.
Codein
EVO
/
EVO
/
EVO
/
EVO
100 µg/ml
250 µg/ml
500 µg/ml
icatibant

Figure 3. Histamine concentrations in dISF collected from *ex vivo* human skin experiments. EVO756 (EVO) was injected prior to injection at the same site with codeine or icatibant. Hollow microneedles were utilized to extract dISF from the injection sites from which histamine levels were quantified.

Phase 1 Healthy Volunteer icatibant Skin Challenge

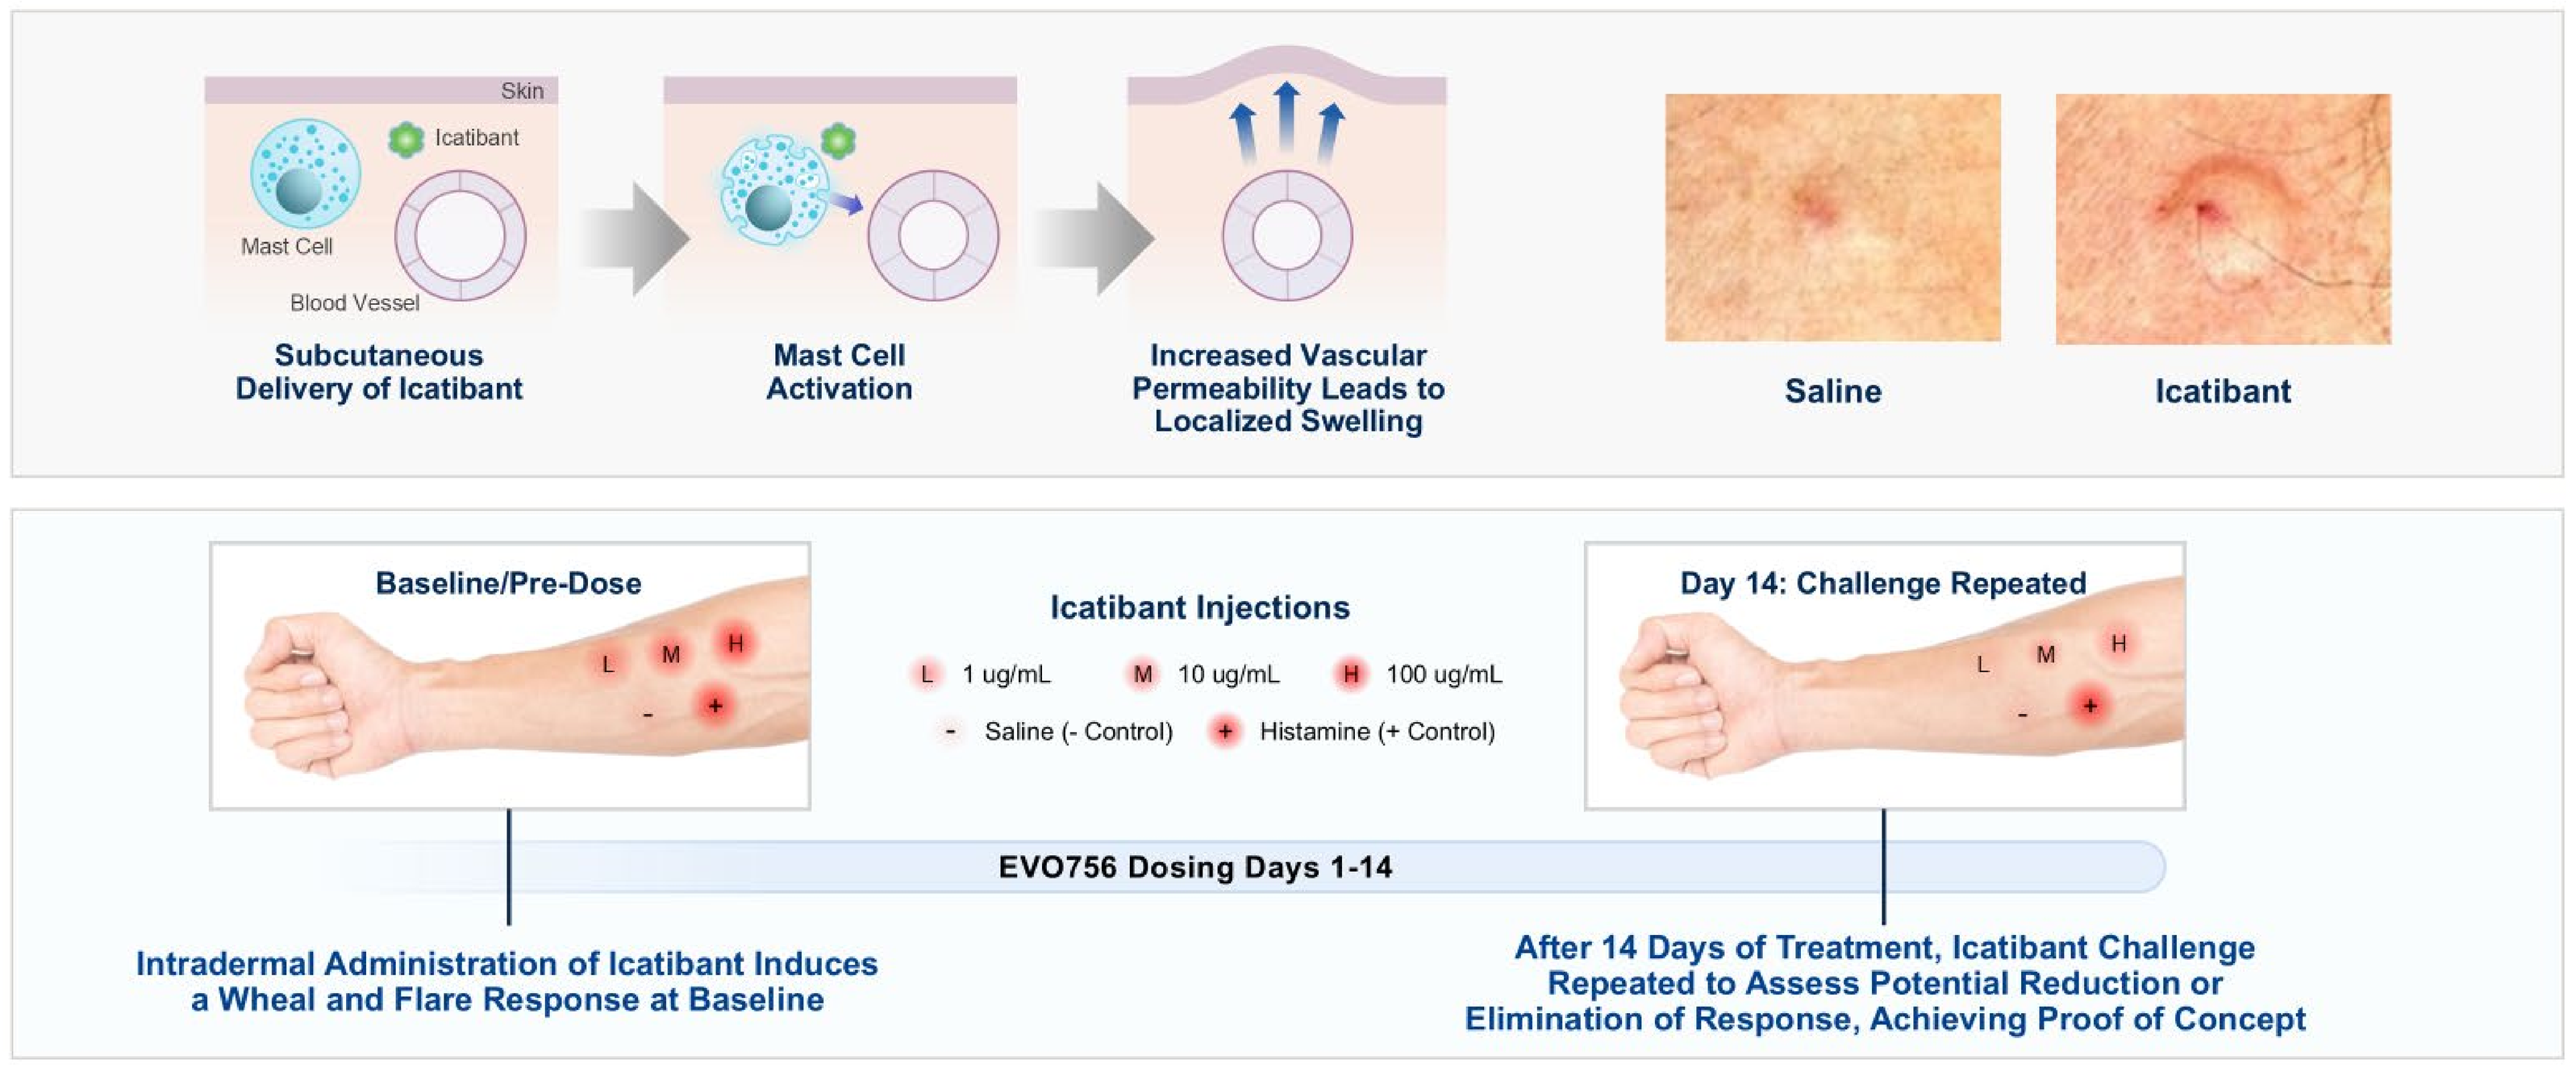
Skin
Icatibant
Mast Cell
Blood Vessel
Subcutaneous Delivery of Icatibant
Mast Cell Activation
Increased Vascular Permeability Leads to Localized Swelling
Saline
Icatibant
Baseline/Pre-Dose
Icatibant Injections
1 ug/mL
10 ug/mL
100 ug/mL
Saline (- Control)
Histamine (+ Control)
Day 14: Challenge Repeated
EVO756 Dosing Days 1-14
Intradermal Administration of Icatibant Induces a Wheal and Flare Response at Baseline
After 14 Days of Treatment, Icatibant Challenge Repeated to Assess Potential Reduction or Elimination of Response, Achieving Proof of Concept

Figure 4. Phase 1 healthy volunteer skin challenge study design with oral EVO756. In brief, the wheal response in healthy volunteers was determined at baseline in response to saline, histamine, or 1, 10, and 100ug/mL of an intradermal injection of icatibant. Healthy volunteers were given 10, 30, 100, 240mg of EVO756 BID or 500mg QD for 14 days. After 14 days or oral EVO756, the skin challenge was repeated as performed at baseline, and reduction in wheal size was noted

EVO756 inhibition of icatibant responses in primary human mast cells *in vitro* correlates with Phase 1 healthy-volunteer skin challenge clinical results *in vivo*

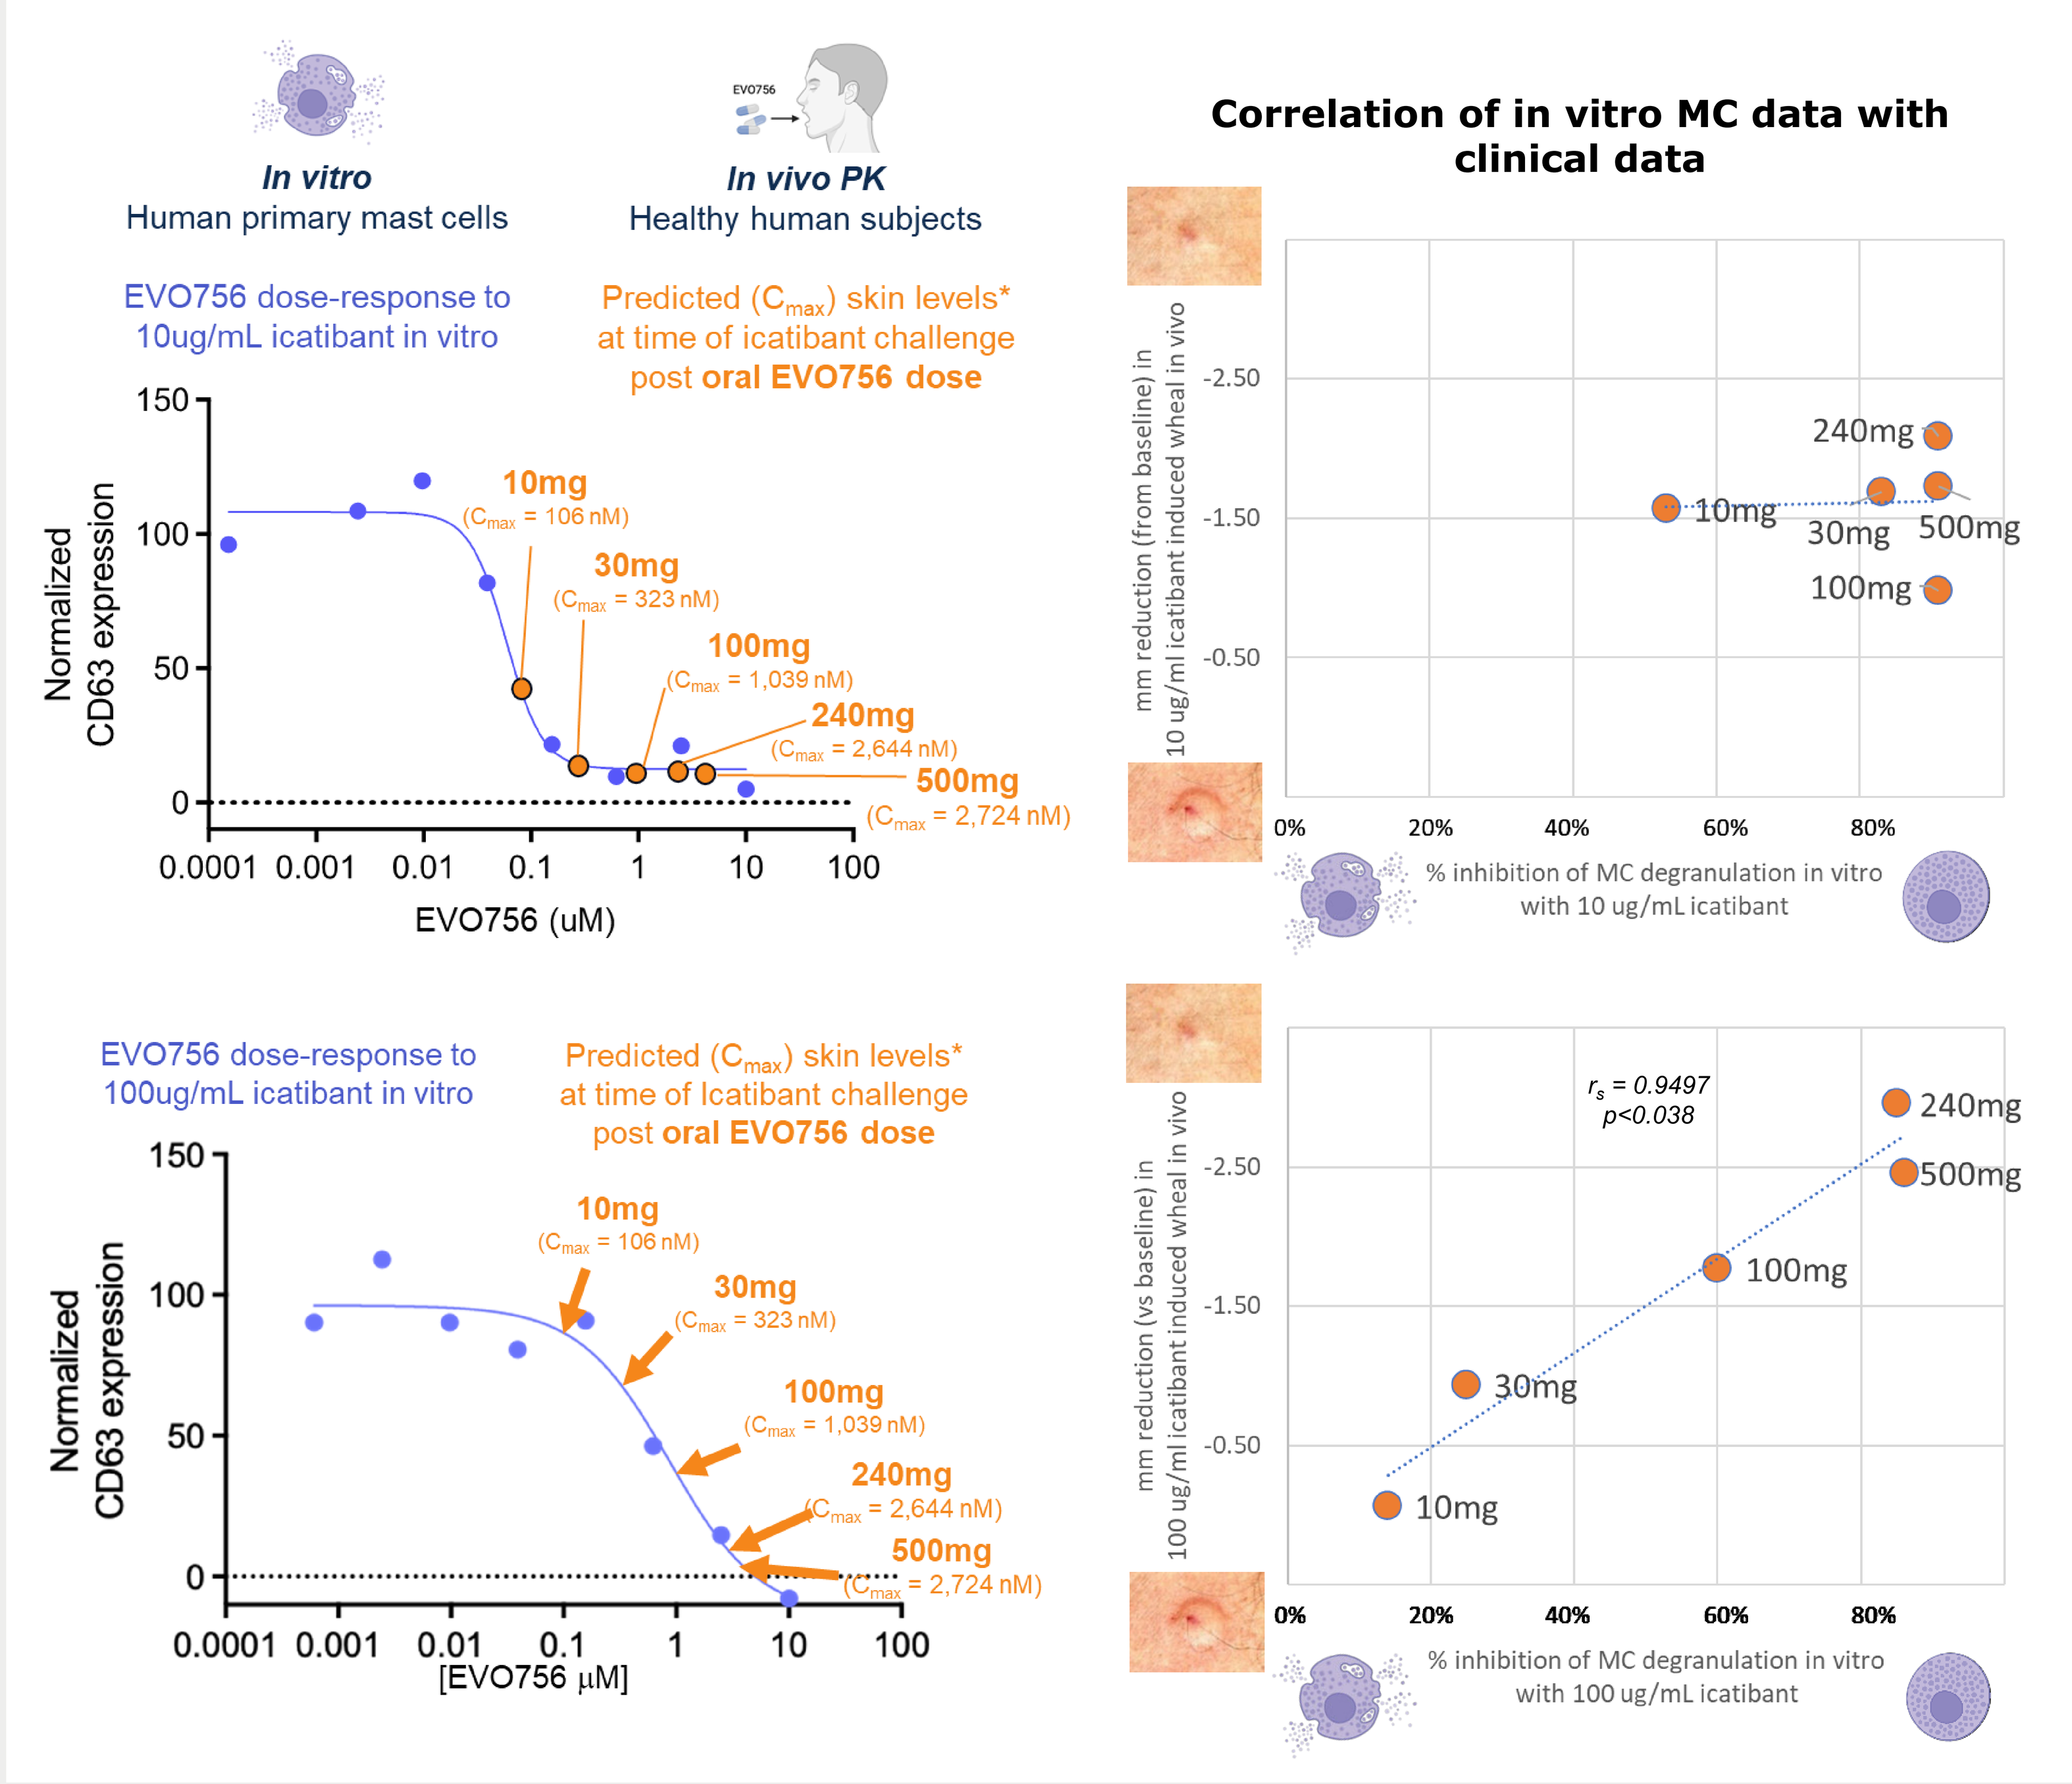
In vitro
Human primary mast cells
In vivo PK
Healthy human subjects
EVO756 dose-response to 10ug/mL icatibant in vitro
Predicted (C_{max}) skin levels* at time of icatibant challenge post oral EVO756 dose
Normalized CD63 expression
EVO756 (uM)
10mg (C_{max} = 106 nM)
30mg (C_{max} = 323 nM)
100mg (C_{max} = 1,039 nM)
240mg (C_{max} = 2,644 nM)
500mg (C_{max} = 2,724 nM)
EVO756 dose-response to 100ug/mL icatibant in vitro
Predicted (C_{max}) skin levels* at time of Icatibant challenge post oral EVO756 dose
[EVO756 μM]
Correlation of in vitro MC data with clinical data
mm reduction (from baseline) in 10 ug/ml icatibant induced wheal in vivo
% inhibition of MC degranulation in vitro with 10 ug/mL icatibant
r_s = 0.9497
p<0.038
mm reduction (vs baseline) in 100 ug/ml icatibant induced wheal in vivo
% inhibition of MC degranulation in vitro with 100 ug/mL icatibant

Figure 5. (Left) EVO756 dose-response curves (blue lines) in phsMCs stimulated with 10 ug/mL (upper) or 100 ug/mL (lower) icatibant, as determined by inhibition of phsMC degranulation via flow cytometry for CD63 cell surface expression (normalized CD63 MFI). Orange arrows point to the predicted maximum concentration (C_{max}) of EVO756 in the skin, based on preclinical tissue distribution studies in rat (~50%), known EVO756 free fraction (16%), and Phase 1 human pharmacokinetic data of oral EVO756. (Right) Correlation of reduction in wheal size *in vivo* from the Phase 1 healthy-volunteer skin challenge study with *in vitro* phsMC data with the predicted C_{max}.

Conclusions

1. Icatibant induced MRGPRX2 responses ON MAST CCELLS are similar to endogenous ligands
2. EVO756 inhibits icatibant induced MRGPRX2 responses similarly to other endogenous ligand induced MRGPRX2 responses in vitro
3. EVO756 can also inhibit icatibant induced histamine release in ex vivo human skin
4. Oral EVO756 administration to healthy volunteers results in a significant dose-dependent inhibition of icatibant-induced wheals. This data highly correlates to *in vitro* data using phsMCs demonstrating the utility of the phsMC assay and the use of icatibant as a prototypical MRGPRX2 agonist, to evaluate MRGPRX2 target engagement

Acknowledgements

We would like to thank Spring Science for their contributions to work presented in this poster. Some illustrations were created with BioRender.com

Disclosures

JLH, SB, BP, AK, JP, and LRB are employees of, and hold stock in, Evommune.